Next-Generation Liquid-Based Slide Immunocytochemical Staining

High Throughput
Throughput: 32 slides.
Each slide supports both single- and dual-marker detection to meet diverse needs.

Flexible
Compatible with multi-brand preservation solution.
Direct-to-instrument workflow.
Supports single-test loading.
Software settings can start from any step.

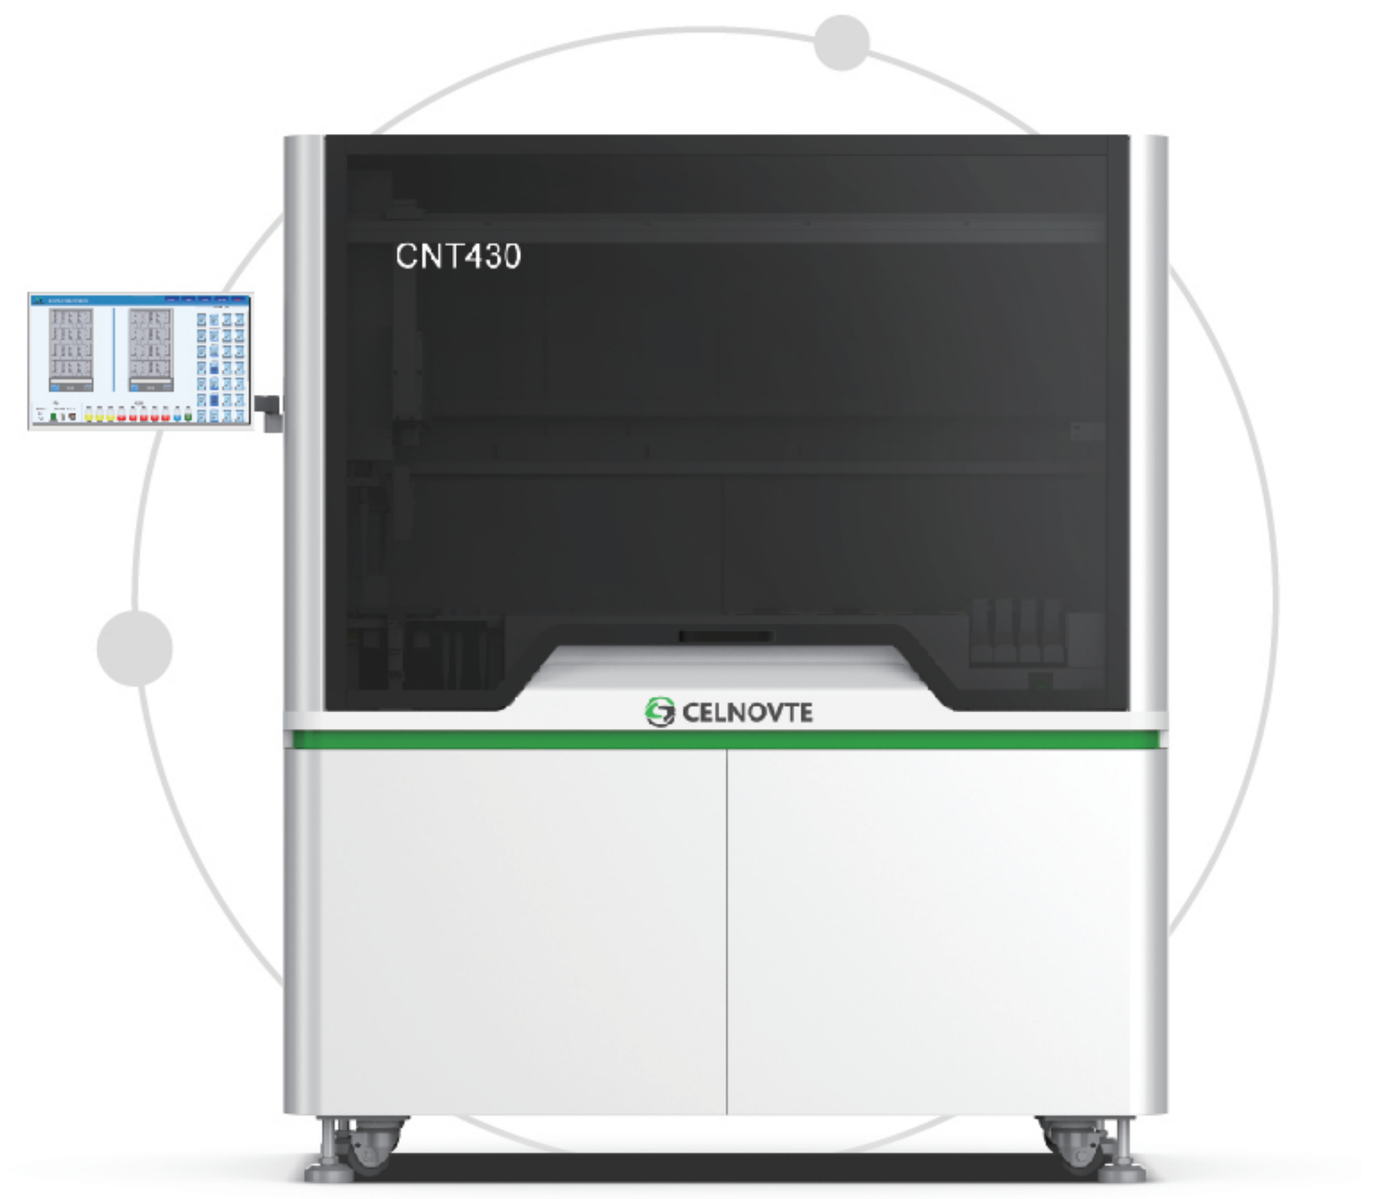
CNT430

High-Sensitivity Reagents
Primary and secondary antibodies optimized for cell samples to enhance detection sensitivity.

Independent Temperature Control
Independent temperature control, flexible setting, meeting the staining requirements of different projects.

Technical Parameters

Main Parameters	
Maximum Throughput	32 (16*2) slides/batch. The intelligent dual-channel structure doubles the efficiency compared with traditional solutions.
Reagent	44 positions, easily meeting diverse experimental needs and making work more efficient and convenient.
Slide Temperature Control	Independent slide trays enable cyclic loading, and each slide has individual temperature control.
Control Terminal	The same instrument completes all steps of fully automatic staining, with a standard integrated computer.
Program	Supports custom program editing to create exclusive solutions as needed.
Monitoring	Real-time monitoring of the experimental process, full-process grasp of dynamic changes, escorting your detection.
Application	Multi-functional, suitable for gynecological and non-gynecological cytology samples, supporting detection items such as ICC/Pap/double staining.
Slide Preparation	Centrifugal enrichment/sedimentation slide preparation.
Software	Supports LIS/HIS.
Consumable Preparation	Supports direct loading of cell preservation vials of different specifications and sizes, with modular loading.
Independent Drip Staining	Each sedimentation chamber has independent drip staining. A single slide can be set with two sedimentation chamber positions to prevent contamination.
Environmental Requirements	
Dimensions	1450mm*850mm*1700mm
Maximum Power	1500VA

TECHNOLOGY
CREATES FUTURE

INTEGRITY
BONDS PARTNERSHIP

Celnovte Biotechnology Co., Ltd.
Tel: +1-4012094076
Email: info@celnovte.com
Website: www.celnovte.com
Address: 9430 Key West Ave, Rockville, MD, 20850

CELNOVTE

CNT430

Fully automatic immunocytochemical stainer

Direct-to-Instrument Analysis of Cell Samples

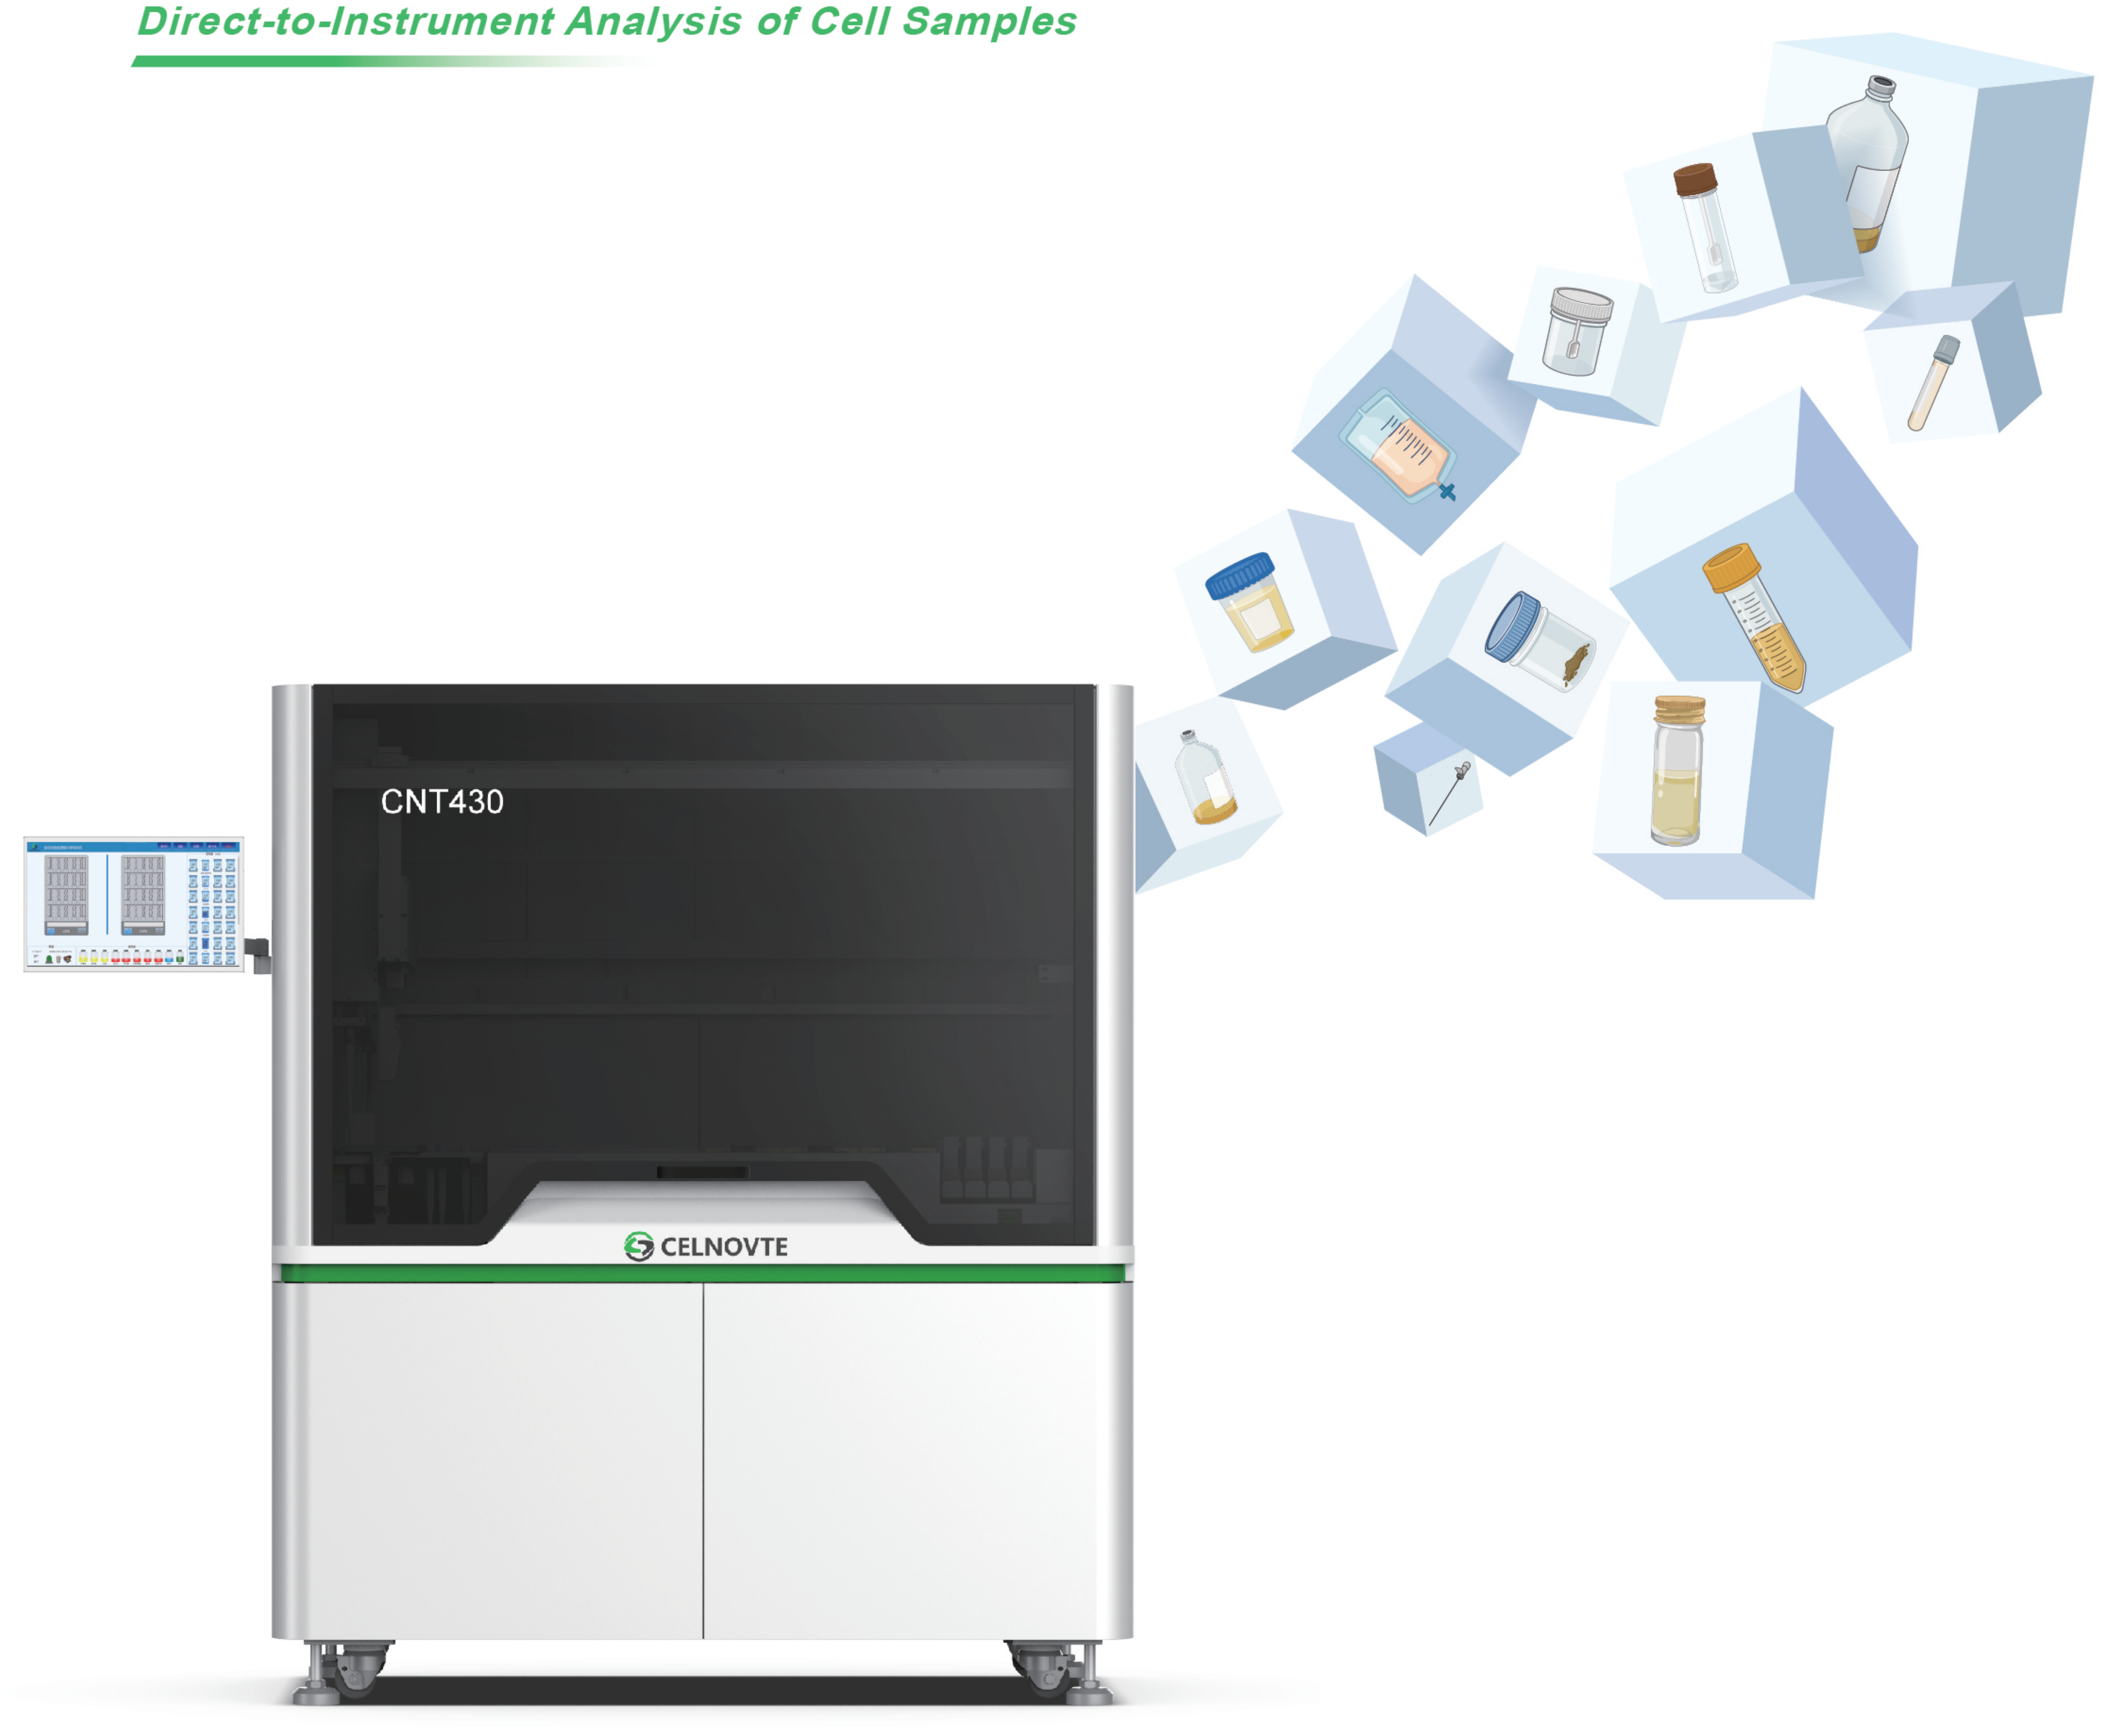
CNT430
CELNOVTE

p16/Ki-67 Patient Triage Optimized

Yellow & Blue Contrast — Instant Staining, Accurate Diagnosis

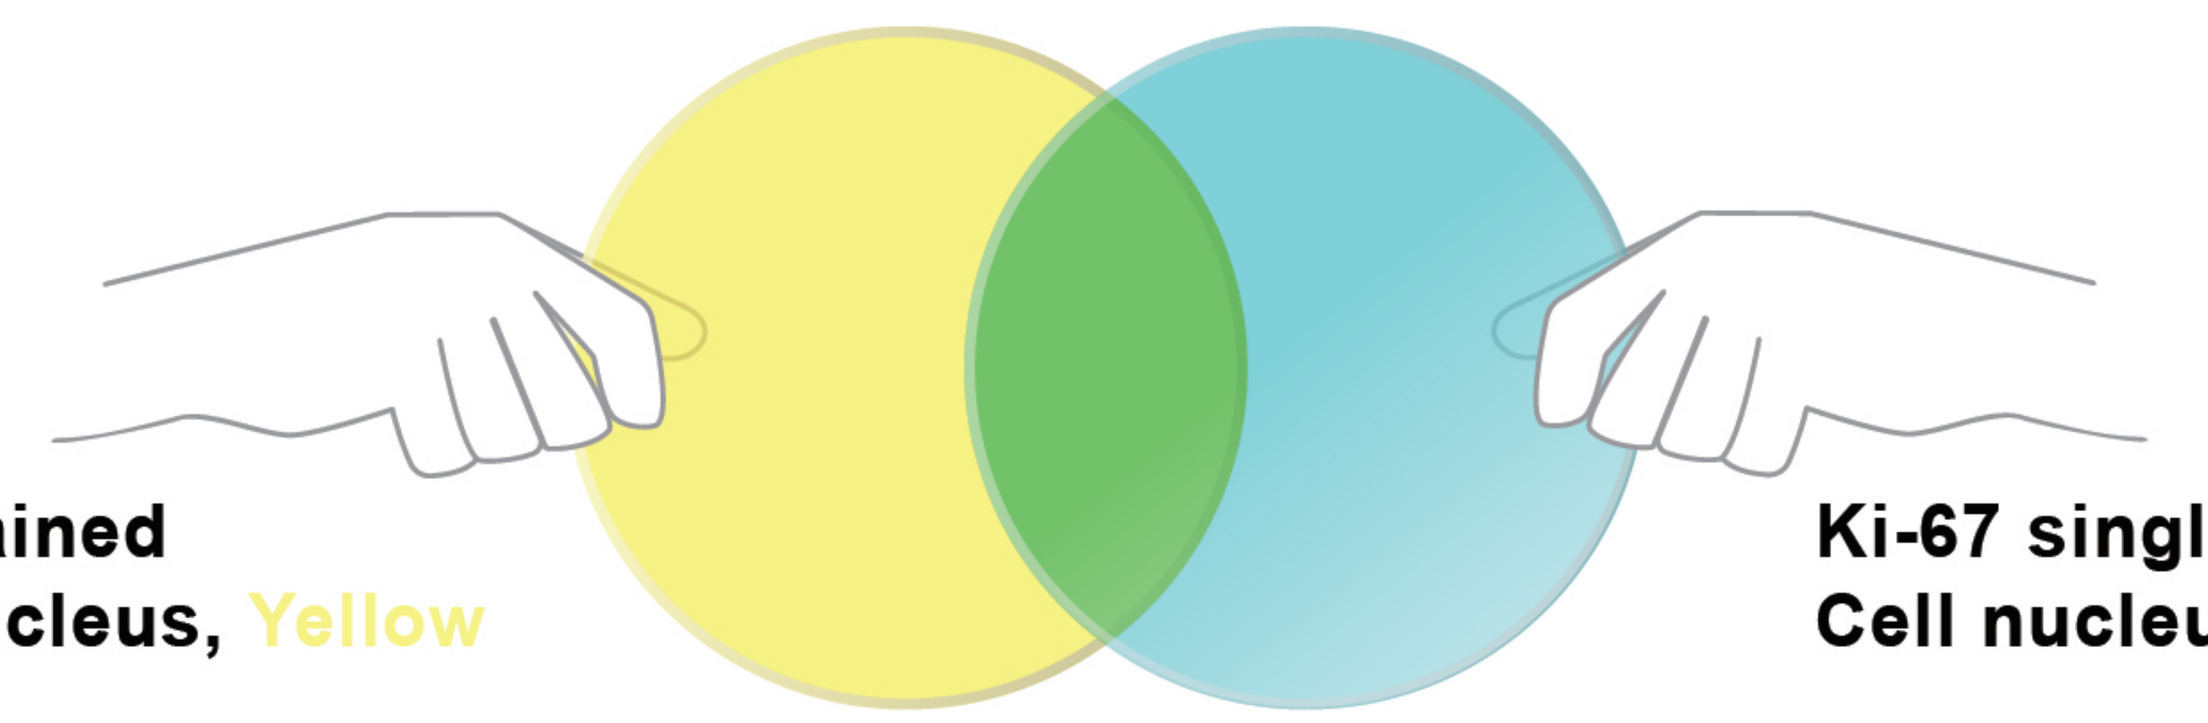

p16 single stained
Cytoplasm/nucleus, Yellow

Ki-67 single stained:
Cell nucleus, blue

p16/Ki-67 Dual stained, Green

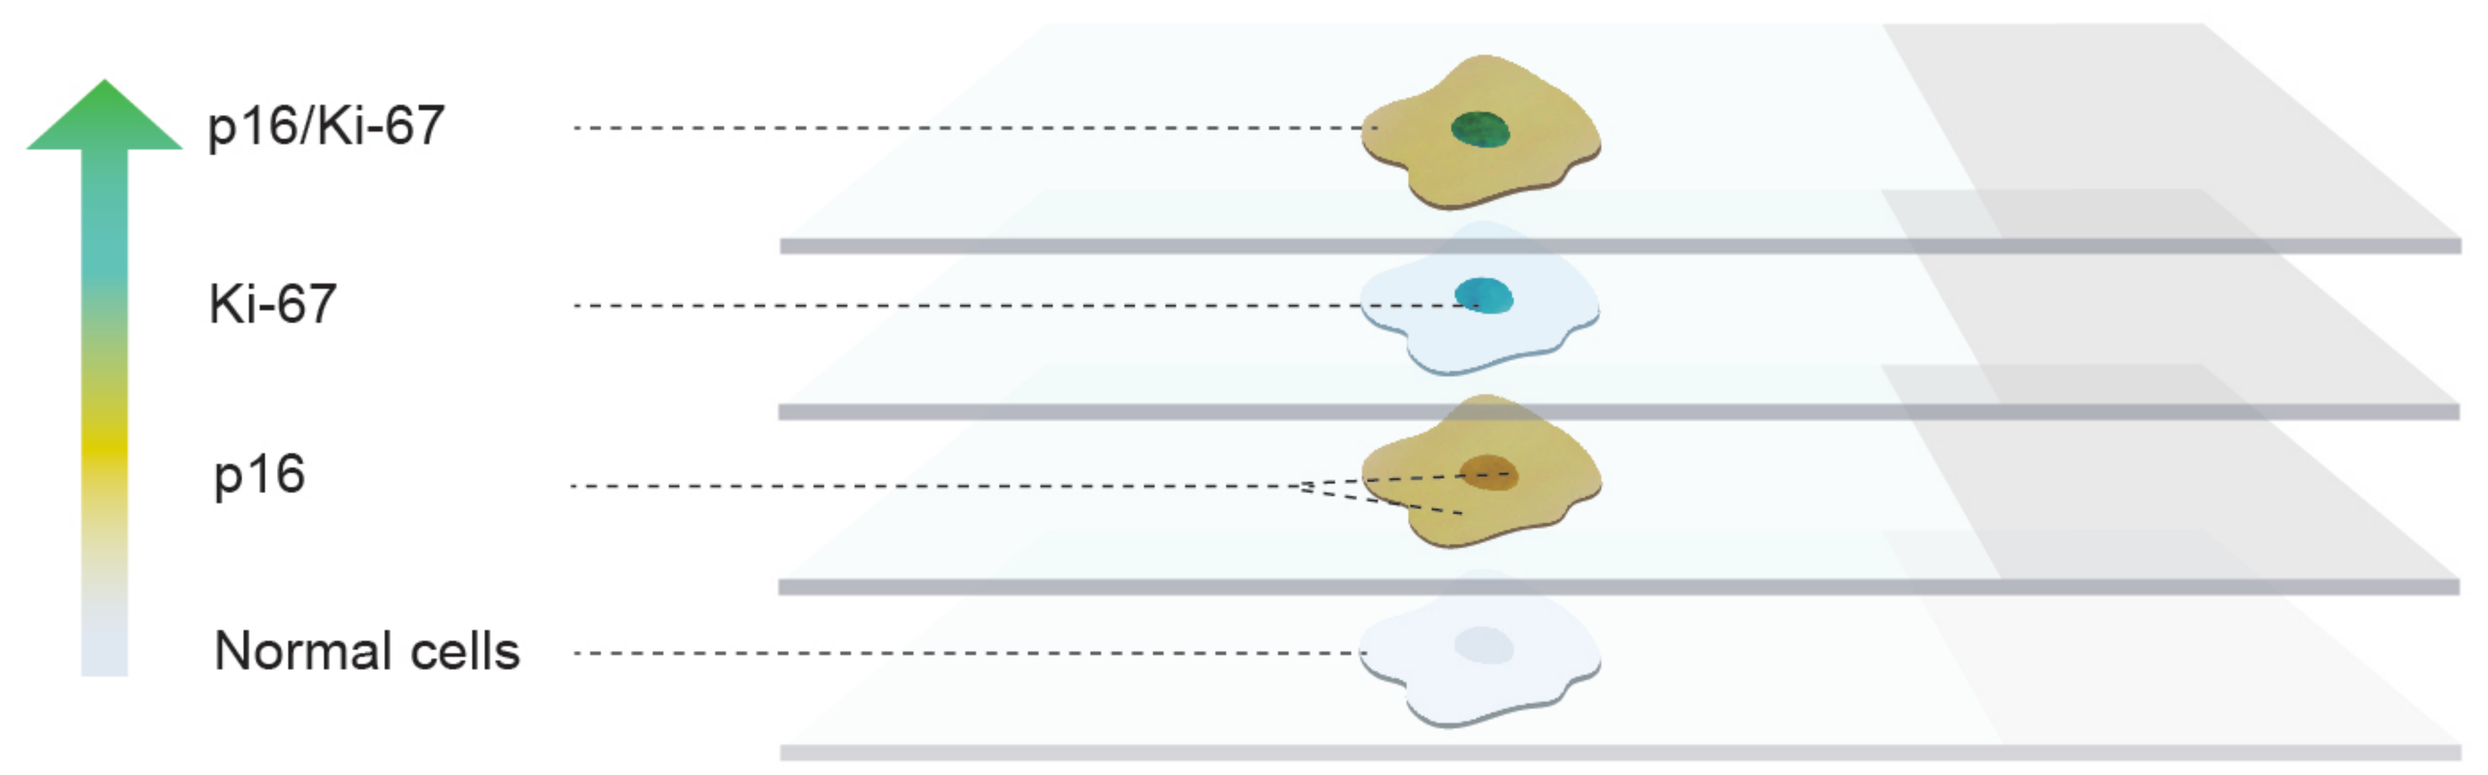
p16/Ki-67
Ki-67
p16
Normal cells

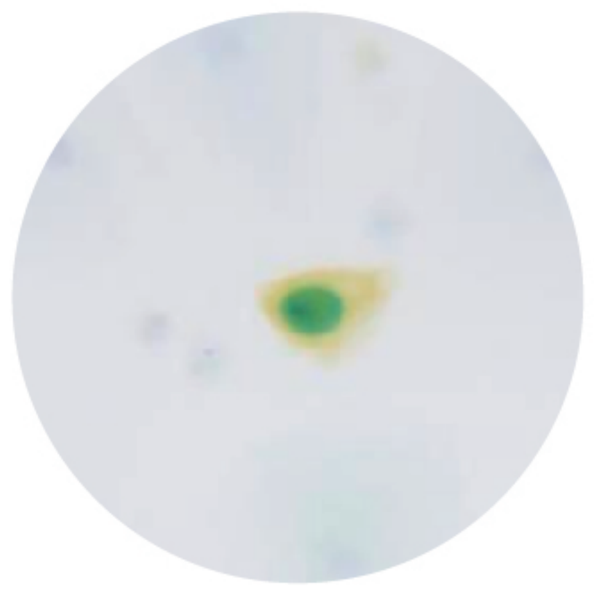

Dual-stained positive cells

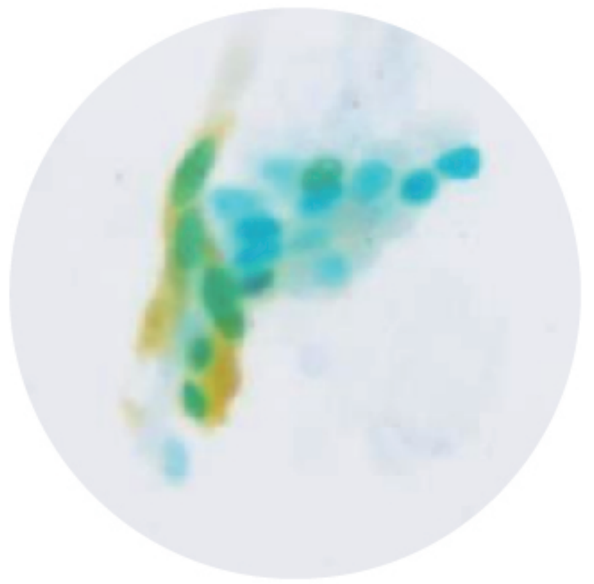

Clusters of dual-stained positive cells

Rapid Detection, Immunocytochemical Staining Ready in 3 Hours

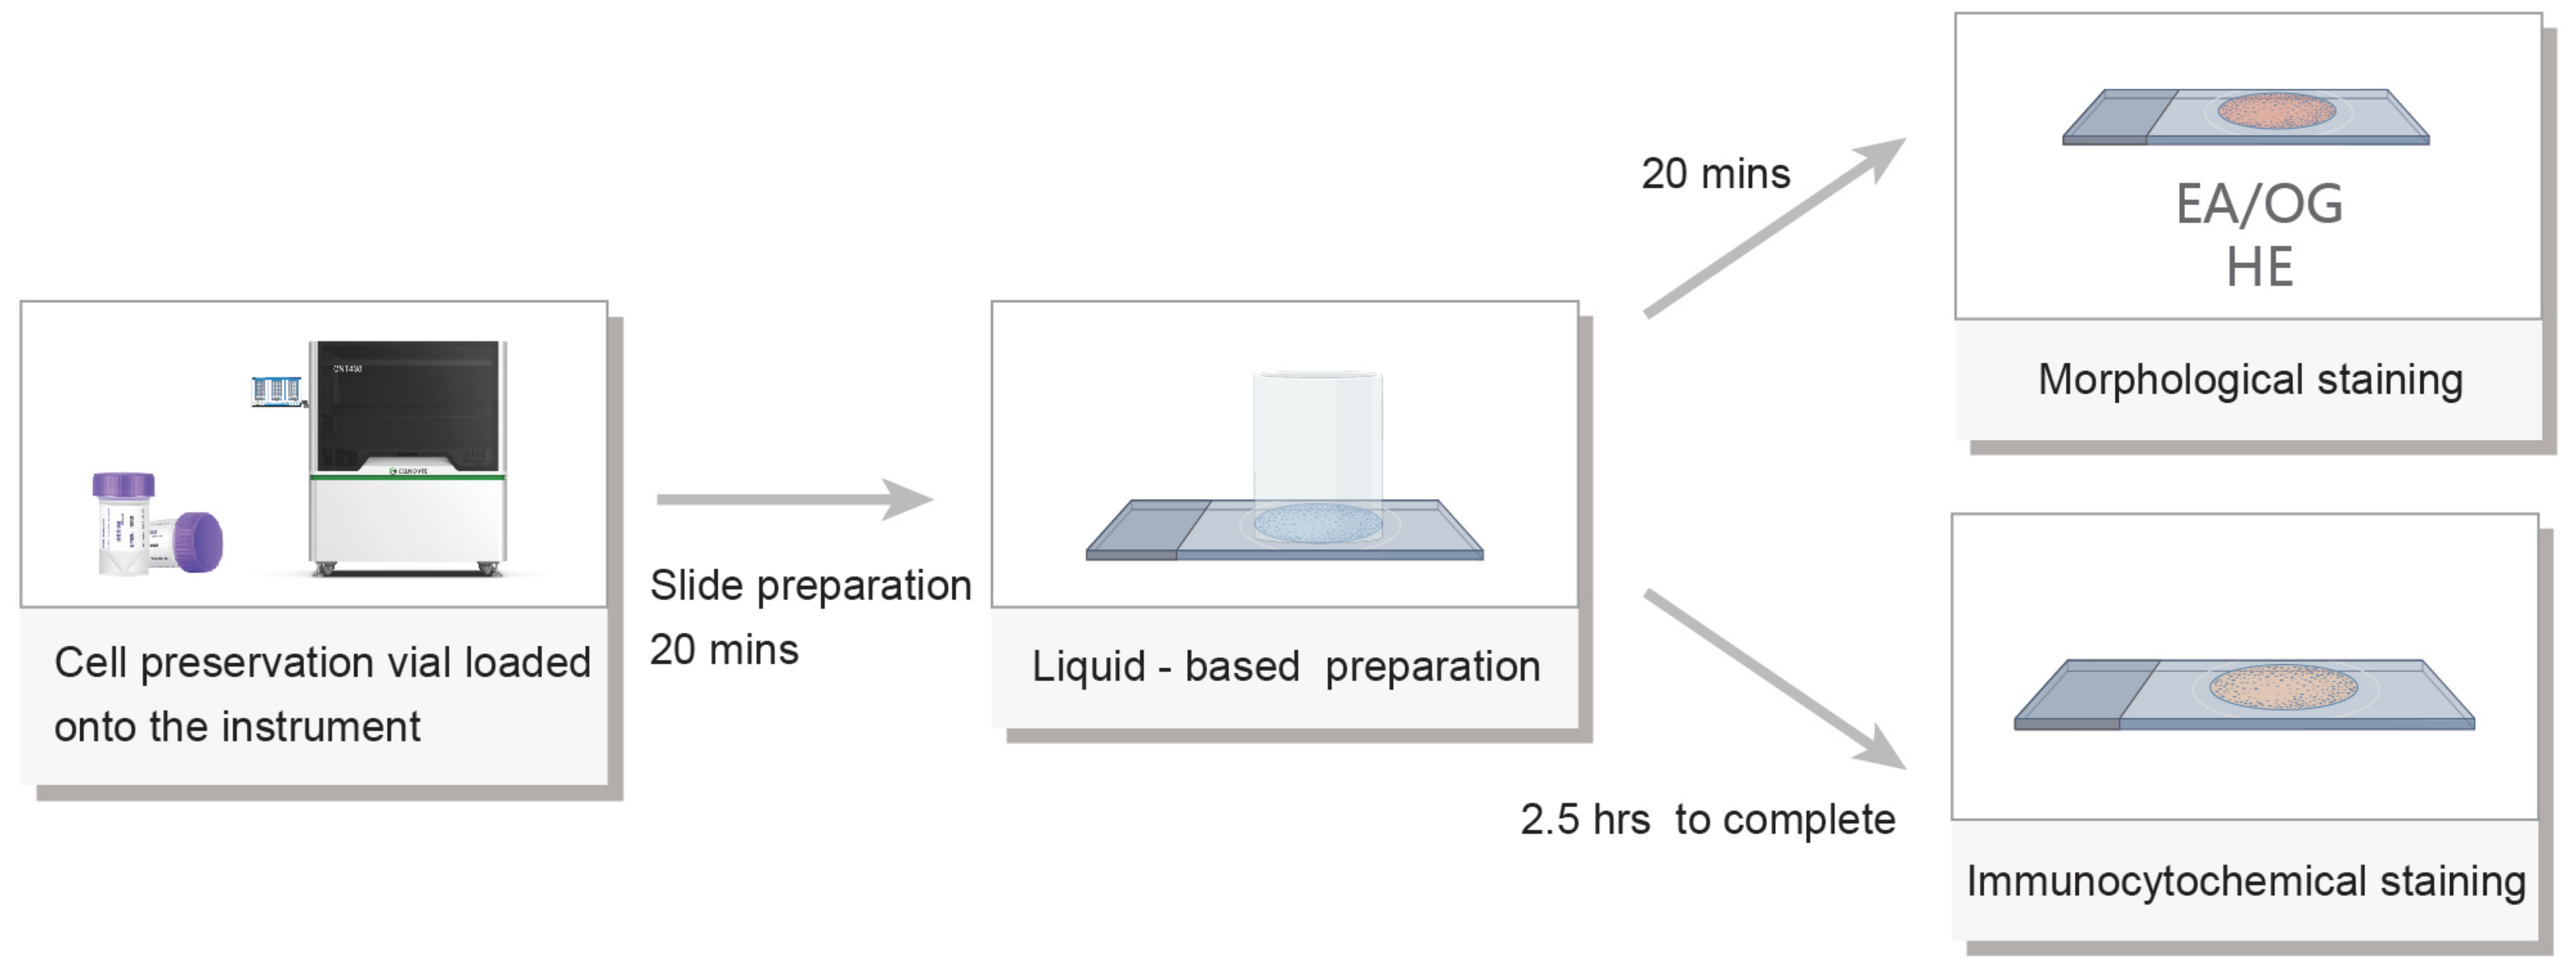
Cell preservation vial loaded onto the instrument
Slide preparation
20 mins
Liquid - based preparation
20 mins
EA/OG
HE
Morphological staining
2.5 hrs to complete
Immunocytochemical staining

ICC Assists in Precise Diagnosis of Non-Gynecological Cellular Pathology

Recommended Applications

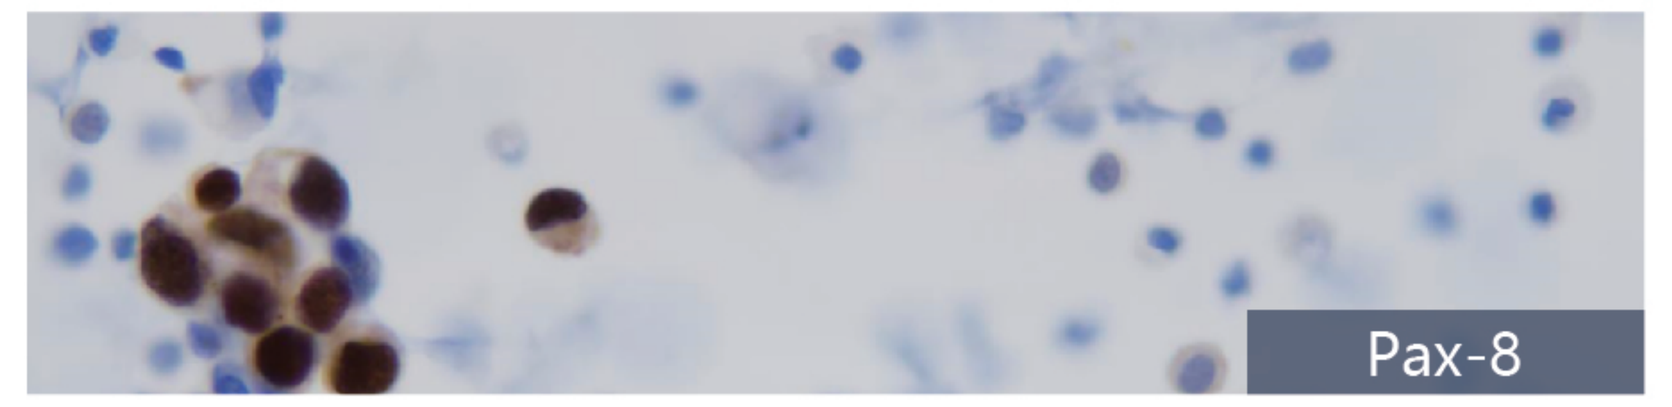
Pax-8

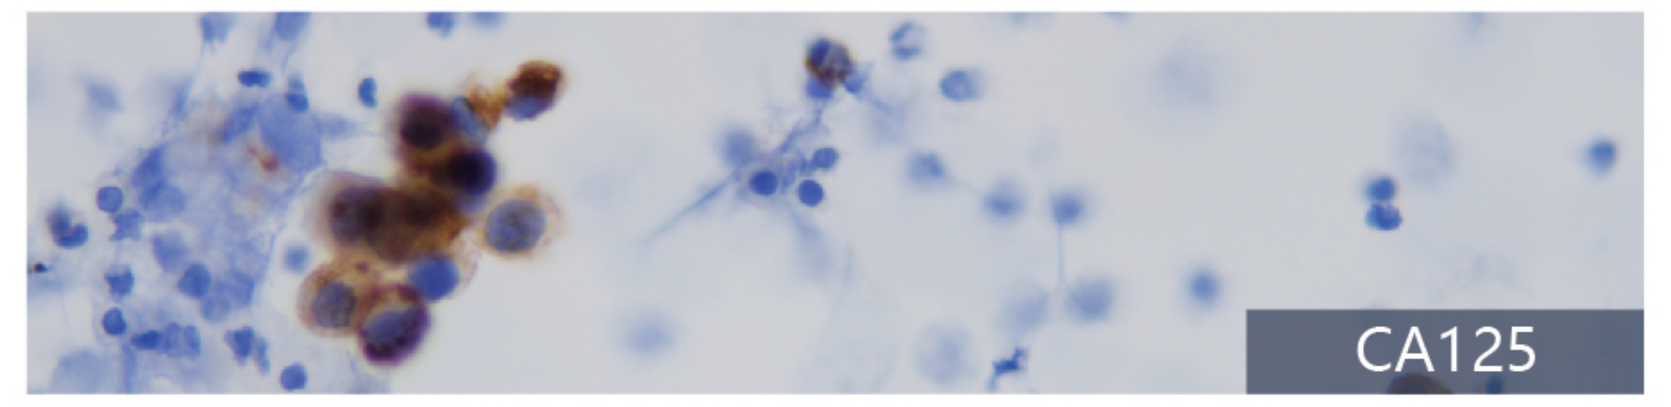
CA125

Ascites Markers

Mesothelium: Calretinin, Desmin, WT-1, CK5/6

Prostate: PSA, CK7

Uterus, Ovary: ER, PR, p16, Pax-8, WT-1, p53, CA125

Digestive Tract: STAB2, CDX-2, CK20, CK

Lung, Urinary Tract: CK7, CK20, GATA-3, p53

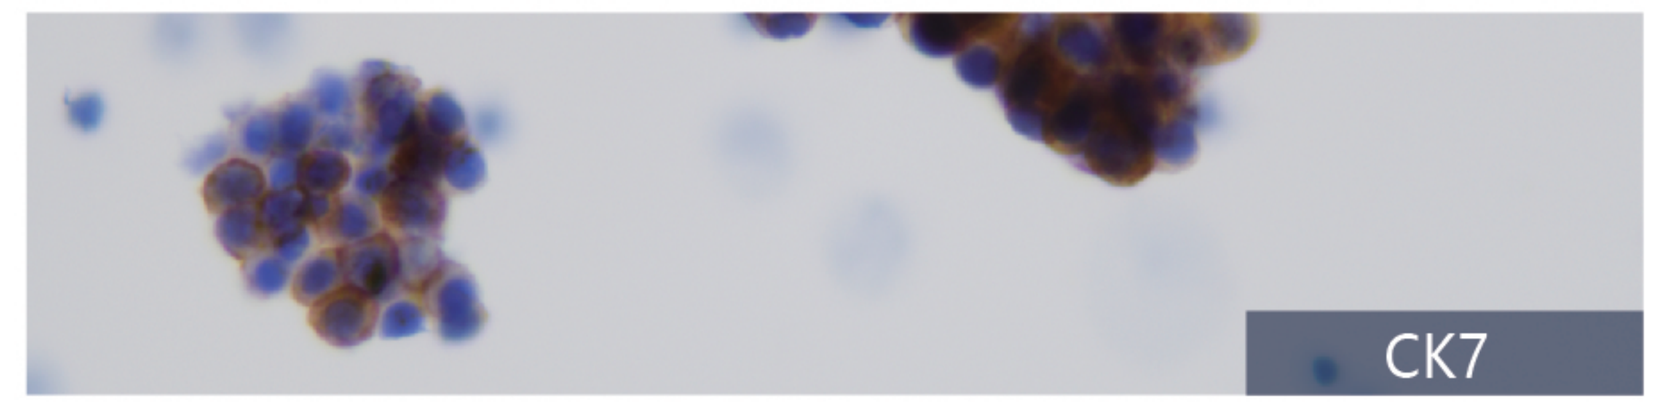
CK7

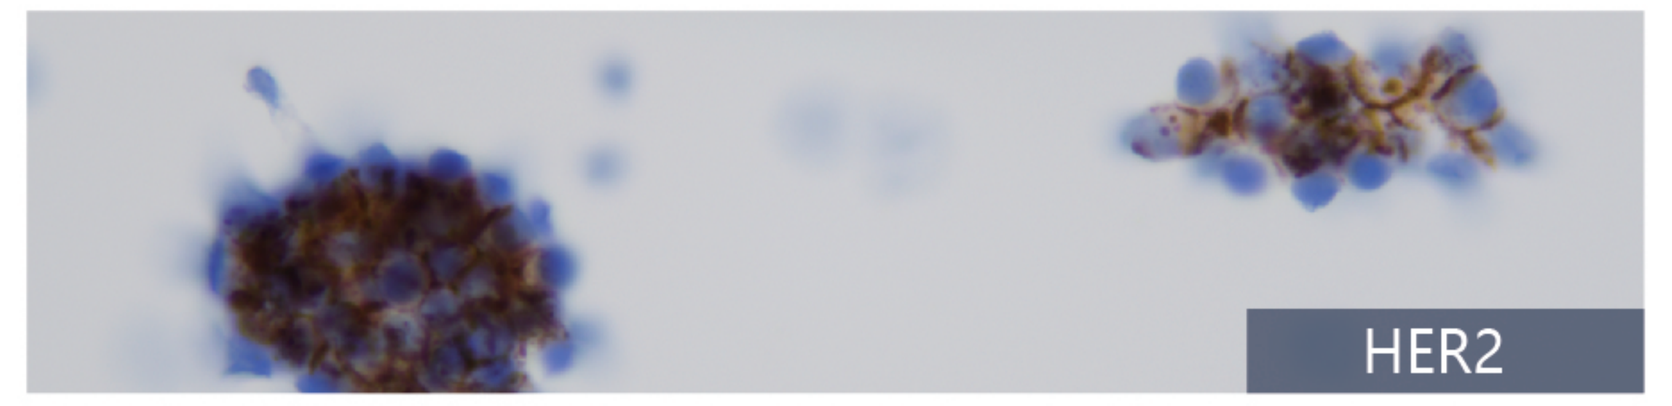
HER2

Pleural Effusion Markers

Mesothelioma: Calretinin, Desmin, WT-1, CK5/6

Lung Squamous Cell Carcinoma: p40, CK5/6

Lung Adenocarcinoma: CK7, TTF-1, EMA, NapsinA

Small Cell Lung Cancer: CgA, CK, TTF-1, CD56, CD45(LCA), Ki-67, NapsinA, Syn, INSM1

Breast Cancer: GATA-3, ER

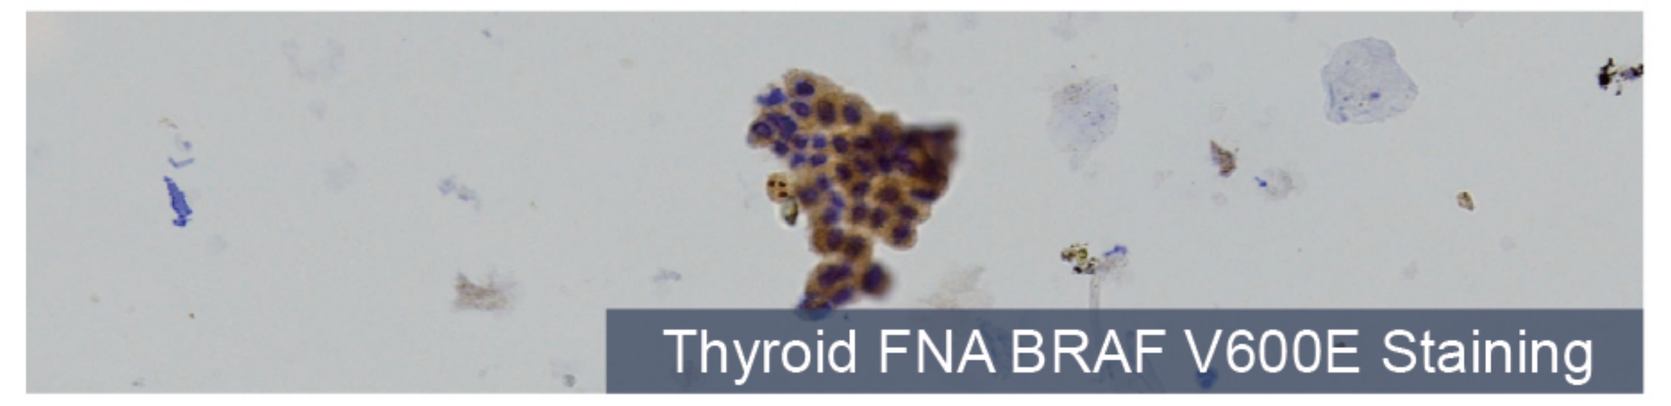
Thyroid FNA BRAF V600E Staining

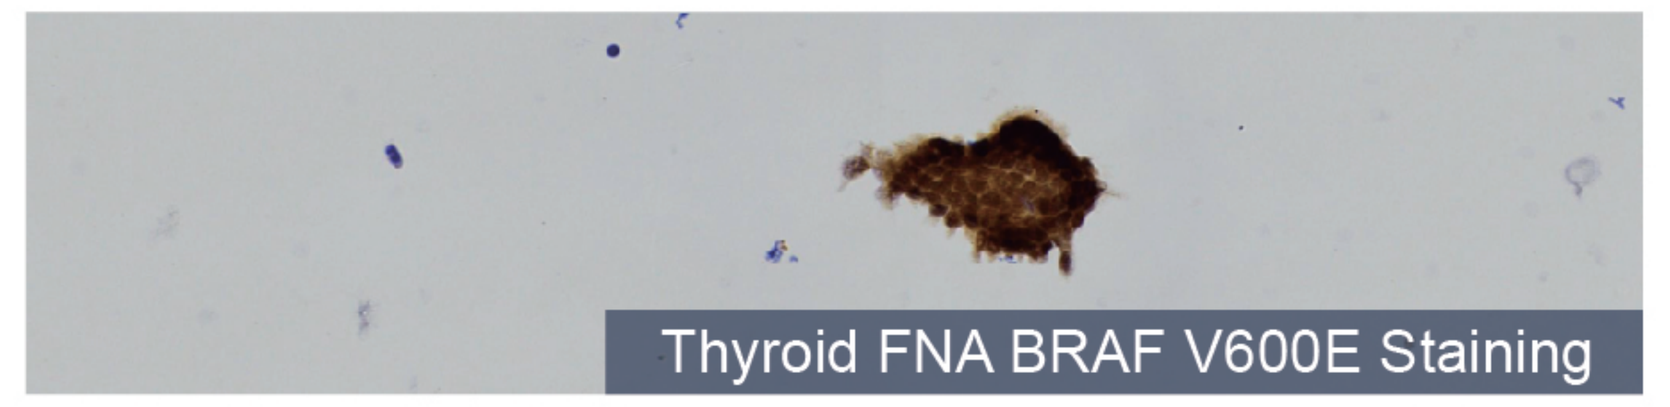
Thyroid FNA BRAF V600E Staining

Aspirated Cells

Aspirated cells fine-needle aspiration of thyroid cells, breast aspiration, aspirated cells from other body cavity organs such as EBUS-TBNA, etc.

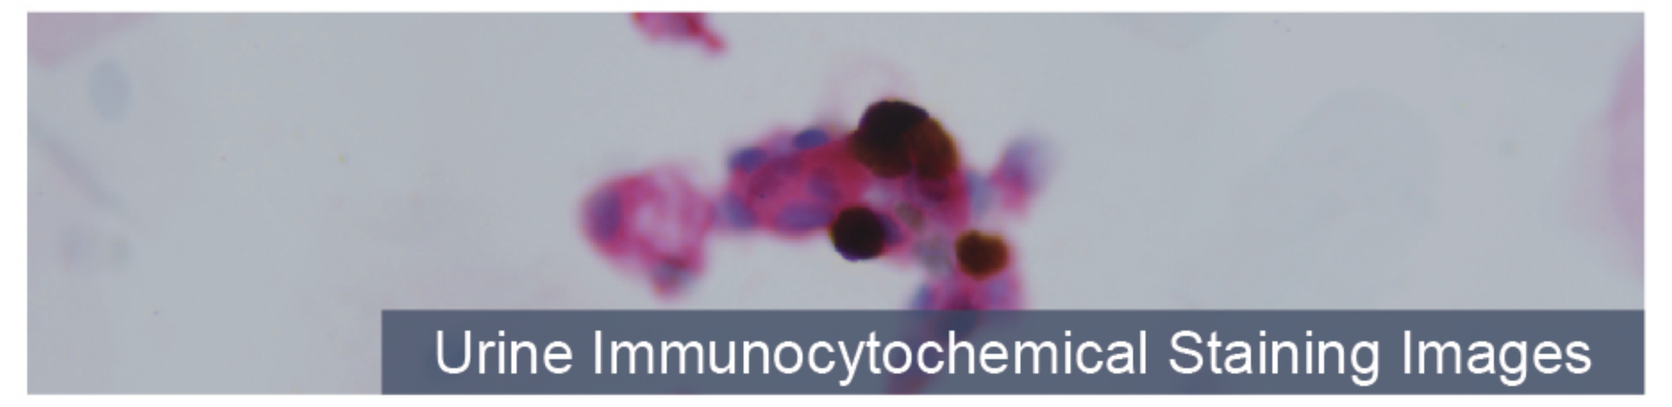
Urine Immunocytochemical Staining Images

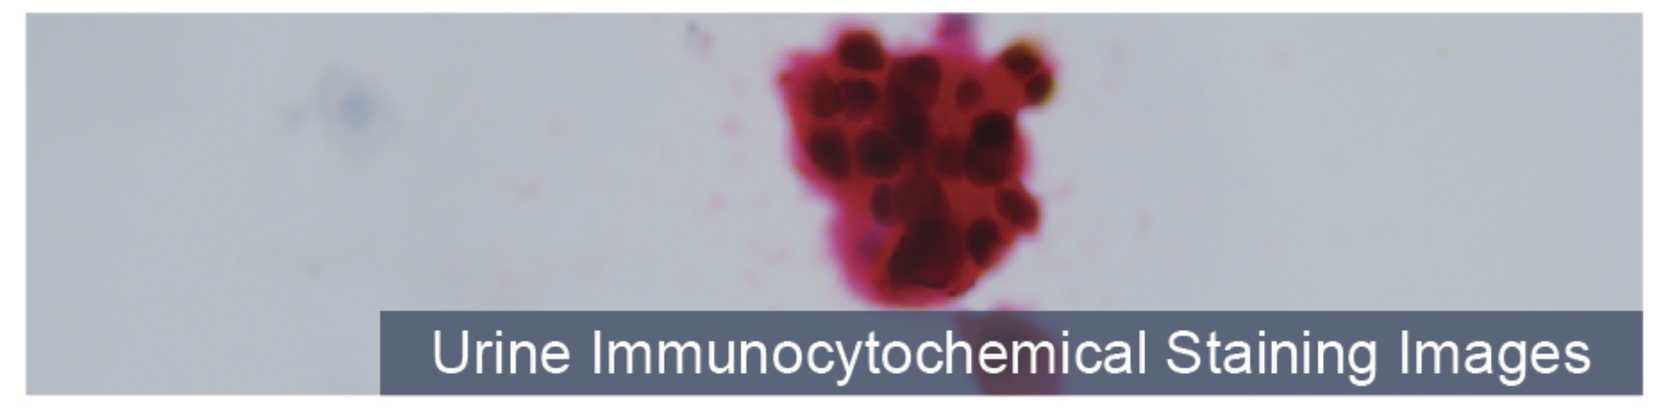
Urine Immunocytochemical Staining Images

Other Cellular Pathology Samples

Other cellular pathology samples cerebro-spinal fluid, urine, bronchial brushing specimens, lung lavage fluid, etc.

p16+TCT Accurate Cervical Cancer Screening

p16 combined with TCT — dual markers, one sample, clear results

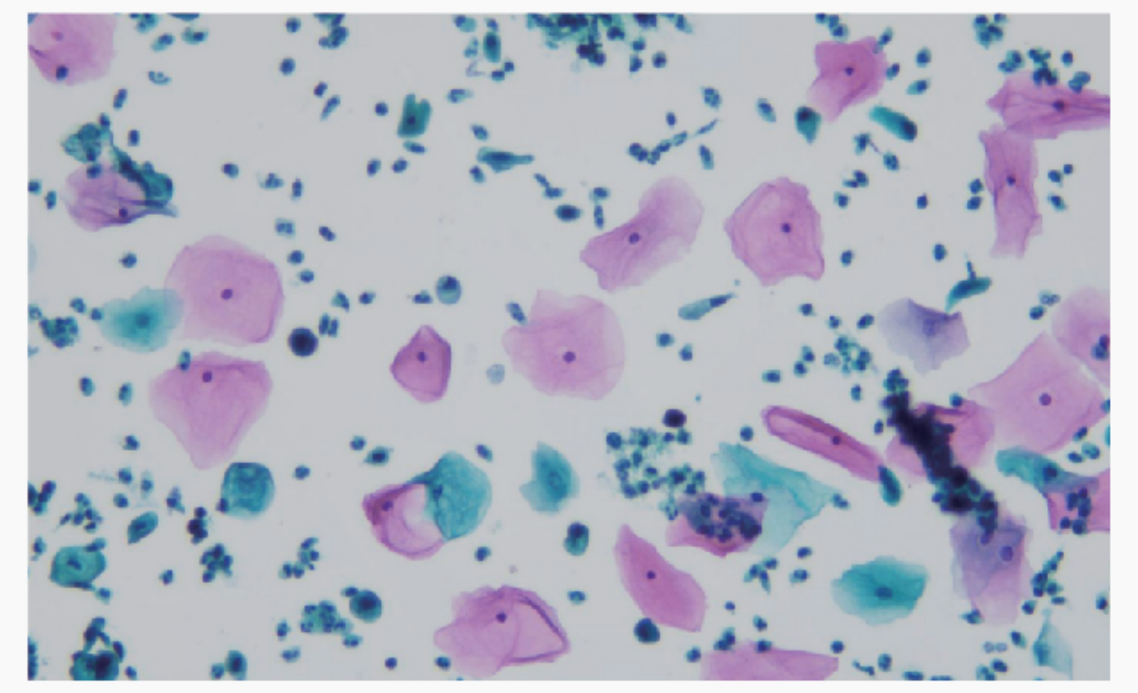

Pap staining

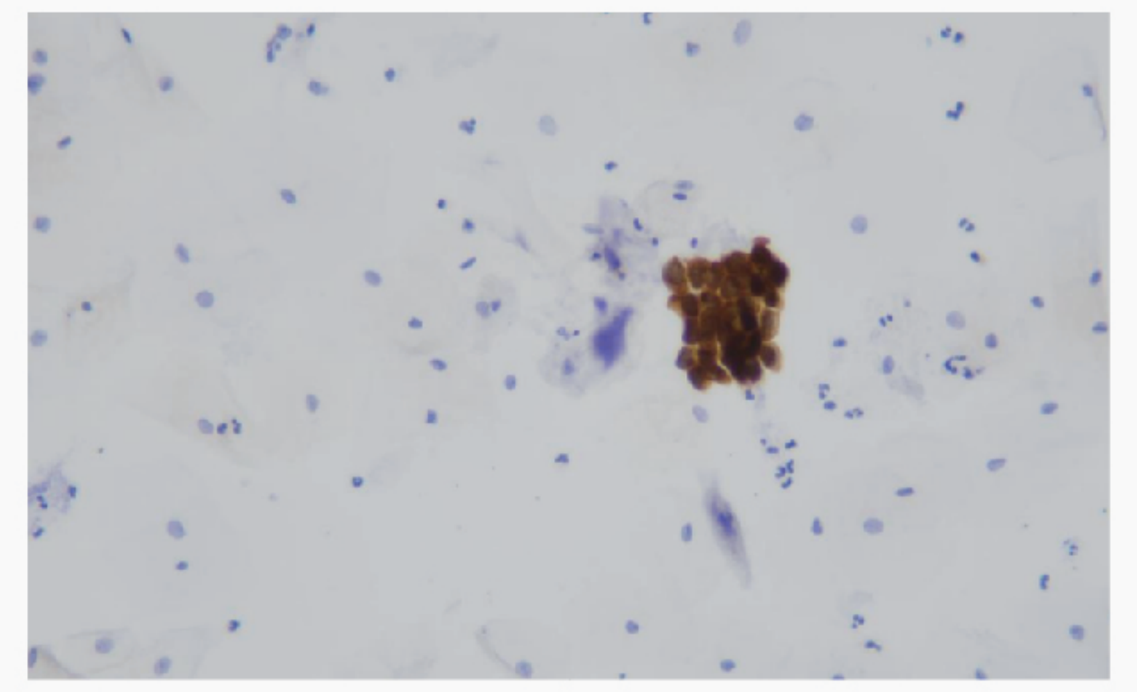

p16 staining

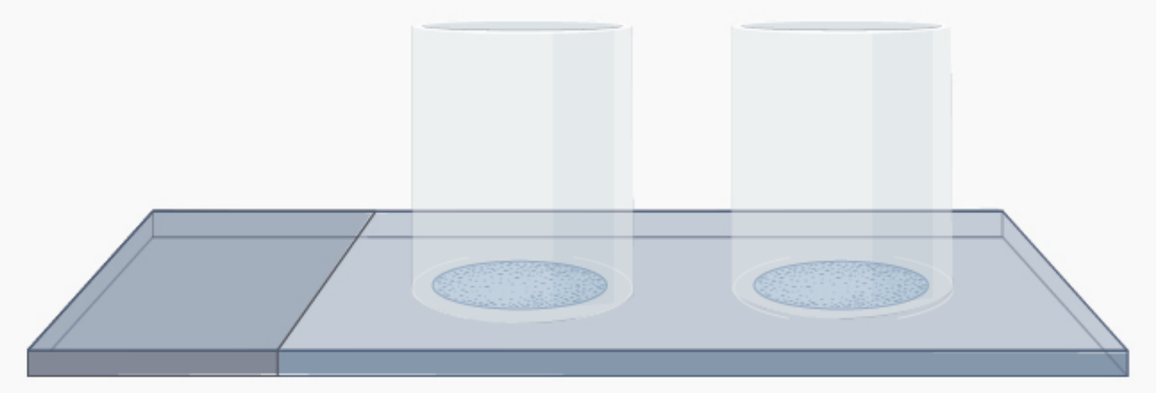

Your choose of single or dual chamber mode

Clinical Value	Implementation ScenariosImplementation Scenarios
High expression of p16 indicates persistent HPV infection.	Dual test can be used as a cervical cancer screening program.
p16 staining helps increase the detection rate of high-grade lesion cells and adeno-carcinoma.	Combining p16 with HPV testing can improve the accuracy of screening and diagnosis for precancerous lesions of the cervix.
Two chamber mode in one sample: the experimental process has less steps, easy operation, and the highly readable results.	p16 testing can be used as a triage program for ASC-US.

New Fully Automated Cervical Cell Scanning and Analysis System (AI) Software

- Can realize dual-marker judgment of cell morphology (TCT) and immunocytochemical staining (p16), improving the accuracy of diagnosis results.
- For single cases, the time of dual-marker diagnosis is within 150 - 180 seconds.
- The negative-excluding rate is 68.8%; the negative-excluding accuracy is higher than 98%.
- Verified by clinical detection , the detection rate of CIN III and above cases is 100%.

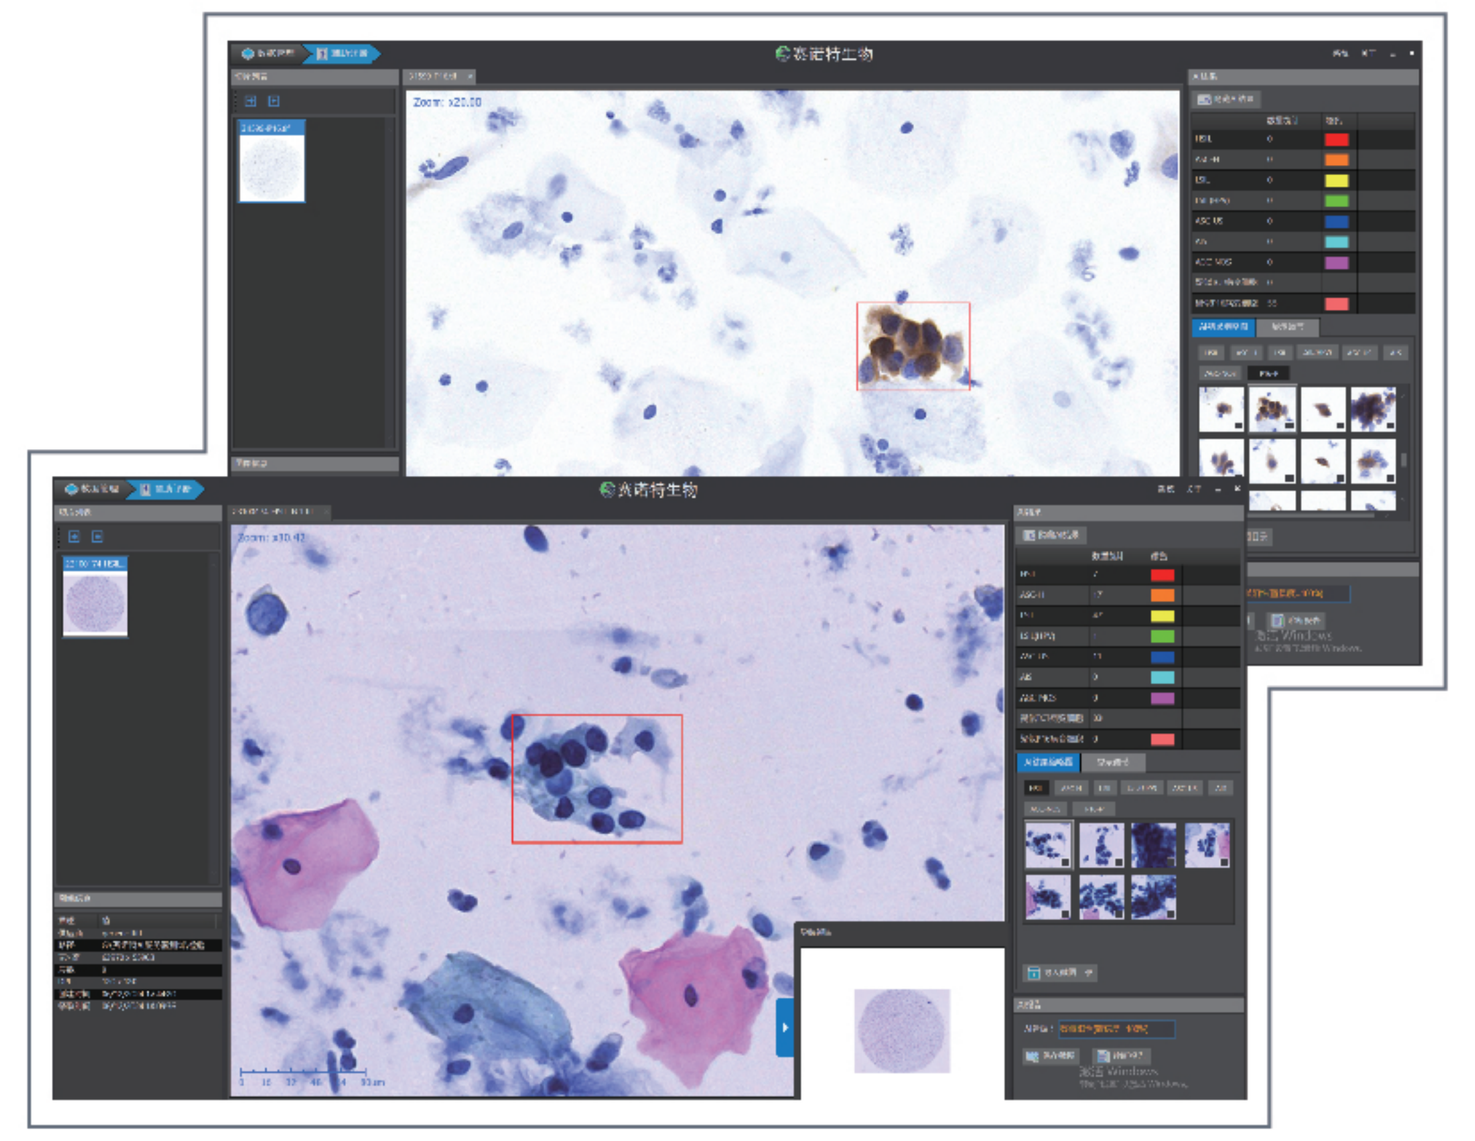